Lowered glucose levels and exogenous insulin requirements in T2DM and T1DM patients treated with oral insulin (ORMD-0801): Phase 2, randomized, placebo-controlled evaluations

For more information: aviva@oramed.com
U.S.:1-646-240-4193; Intl.:+972-2-566-0001
www.oramed.com

[1]Miriam Kidron, Ph.D.; [2]Kenneth Homer; [3]Joel Neutel, M.D.
[1]Oramed Pharmaceuticals, Jerusalem, Israel; [2]Integrium LLC, Cedar Knolls, New Jersey, USA; [3]St. Joseph Hospital, Tustin, CA, USA

PURPOSE

Consensus regarding the convenience of oral insulin, alongside the proposed therapeutic advantages of this delivery route over systemic exposure, have fueled numerous attempts at design of such a formulation. Portally infused insulin brings to more rapid and pronounced suppression of hepatic glucose production and to reduced circulating peripheral insulin levels as compared to systemically administered insulin. Oral insulin deposited directly into the portal vein is expected to have similar salient effects. Oramed Ltd. has developed an oral insulin formulation (ORMD-0801), which harnesses excipients to both hinder proteolysis in the small intestine and enhance translocation of insulin across the gut epithelial lining. Once transported across the gut wall, the insulin is ferried to the hepatic portal vein, mimicking the natural route of endogenous pancreatic insulin, and then subjected to first-pass metabolism in the liver, before being delivered to peripheral sites of action. The PK/PD profile of ORMD-0801 is well suited for the control of fasting blood glucose due to the delayed onset of action. Therefore, Oramed is pursuing the bed-time oral administration of ORMD-0801 for the treatment of elevated fasting blood glucose in adult patients with T2DM. In parallel, the drug has been shown to minimize glucose instability when provided as an adjunct to subcutaneous insulin regimens in T1DM patients.

TYPE 1 DIABETES MELLITUS

Design: In this randomized, double-blind, placebo-controlled, prospective study, 25 adult T1DM patients were outfitted with a blinded continuous glucose monitor (CGM). During the 3-day run-in period, patients were treated with placebo capsules (3 times daily, 45 min before meals) to determine baseline patient insulin requirements. A fresh CGM cannula was then fitted for the following 7-day in-patient treatment period, during which, patients were dosed 3 times daily, 45 min before meals, with either ORMD-0801 (8 mg insulin) or placebo capsules. Study staff administered insulin according to the patient's sliding scale, with adjustments made, as necessary.

Results: On all treatment days, ORMD-0801-treated T1DM patients showed consistently lower fasting plasma glucose (FPG) levels as compared to baseline, peaking at -60.2±63.3 mg/dL on day 7, versus a -10.2±55.7 mg/dL change measured for the placebo cohort at the same time point (Figure 1). Reduced FPG levels directly correlated with reduced rapid-acting insulin requirements, reaching a mean difference of -5.9 mIU/mL insulin intake between active versus placebo-treated patients on day 7 (Figure 2). On day 7 of treatment, an equal number of hypoglycemic events (<60 mg/dL) requiring clinical intervention was reported for each cohort (n=12 per cohort).

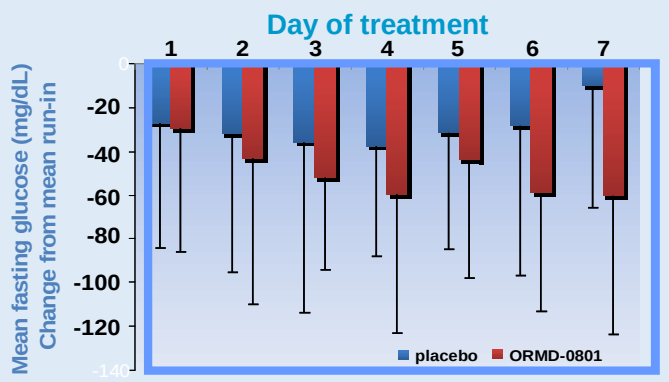
Day of treatment
1
2
3
4
5
6
7
Mean fasting glucose (mg/dL) Change from mean run-in
-20
-40
-60
-80
-100
-120
placebo
ORMD-0801

Figure 1. Mean fasting glucose concentrations. Morning fasting fingerstick glucose measurements were performed and are presented as the mean change from baseline glucose levels. Error bars represent standard deviation.

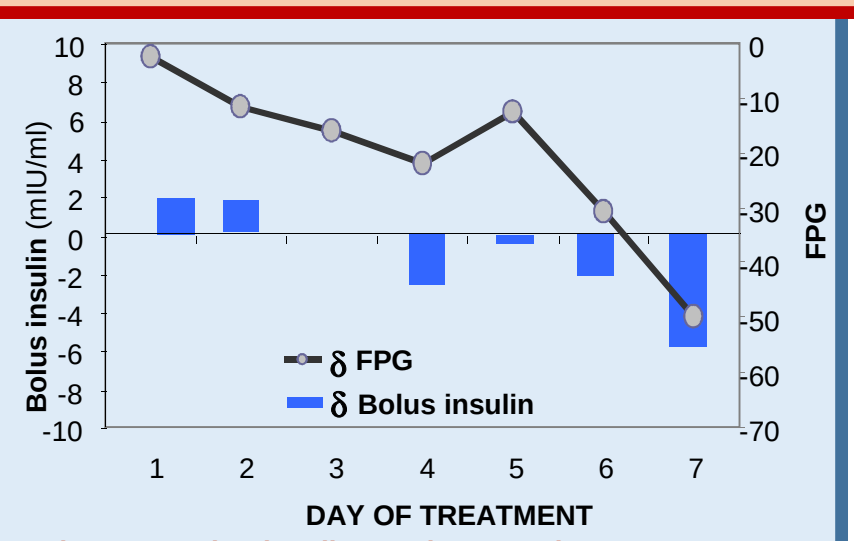
Bolus insulin (mIU/ml)
10
8
6
4
2
0
-2
-4
-6
-8
-10
FPG
0
-10
-20
-30
-40
-50
-60
-70
δ FPG
δ Bolus insulin
1 2 3 4 5 6 7
DAY OF TREATMENT

Figure 2. Bolus insulin requirements in ORMD-0801- vs. placebo-treated T1DM patients. Difference of mean daily bolus insulin requirements of the active versus placebo treatment cohorts plotted against mean FPG levels (bars).

TYPE 2 DIABETES MELLITUS

Design: In this Phase IIb randomized (1:1:1), double-blind, placebo-controlled, multicenter (n=33) study, 192 adult patients with T2DM, participated in a 14-day placebo run-in period, followed by a 28-day treatment period with 16 mg ORMD-0801, 24 mg ORMD-0801 or placebo, self-administered at bedtime. Glucose levels were monitored, via a blinded CGM, during the last 7 days of the run-in and treatment periods.

Results: The active treatment proved safe, well-tolerated and nonimmunogenic, with no serious adverse drug-related events reported. No significant difference in incidence and types of adverse events, including hyper/hypoglycemia, was noted between cohorts. CGM data indicated a significantly smaller change from baseline in nighttime glucose levels in the pooled ORMD-0801 (1.7 mg/dL) as compared to the placebo (13.7 mg/dL, p=0.027) cohort. Mean 24-hour glucose readings remained stable among ORMD-0801-treated patients (mean difference: -0.32 mg/dL), whereas patients receiving placebo demonstrated a mean 13.26 mg/dL change from baseline in these readings (p<0.0001). Similarly, mean change from baseline in fasting (5AM-7AM) and daytime (6AM-10PM) CGM glucose were significantly smaller among ORMD-0801-treated patients as compared to those treated with placebo (-0.4 mg/dL vs. 16.0 mg/dL [p<0.0001] and 0.9 mg/dL vs. 11.9 mg/dL [p<0.001] respectively). In parallel, by the end of the 28-day treatment period, the mean change from baseline in HbA1c levels in the combined ORMD-0801 cohorts (-0.01%) was significantly smaller as compared to the placebo cohort (0.2%; p=0.01), and was projected to show a 0.5% drop from baseline following 12 weeks of treatment.

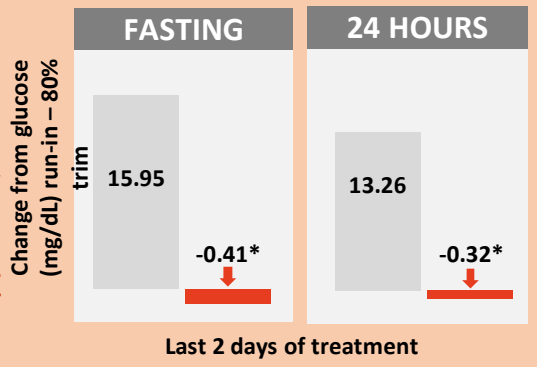
FASTING
24 HOURS
Change from glucose (mg/dL) run-in – 80% trim
15.95
-0.41*
13.26
-0.32*
Last 2 days of treatment

Figure 3. Mean change in blood glucose concentrations: last two days of run-in (gray) vs. last two days of treatment period (orange) Data from the last two days of monitoring that contained at least 80% of the readings were used for the comparative analysis. No significant difference in readings was observed between the two treatment groups and data are therefore presented as a pooled ORMD-0801 treatment cohort.

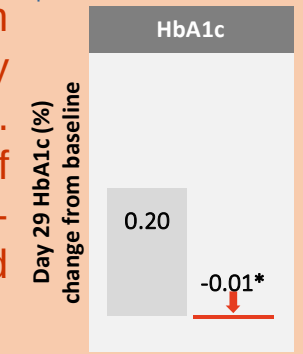
HbA1c
Day 29 HbA1c (%) change from baseline
0.20
-0.01*

Figure 4. Mean change from baseline HbA1c levels following placebo (gray) vs. ORMD-0801 (orange) treatment

CONCLUSIONS

Preprandial ORMD-0801 reduced the exogenous short-acting insulin demands required to maintain euglycemia in T1DM patients and to a greater drop in FPG concentrations, when compared to placebo treatment, seemingly due to improved hepatic insulinization and subsequently normalized gluconeogenesis/glycogenolysis ratios. ORMD-0801 treatment elicited a sustained reduction in mean nighttime, fasting, daytime and 24-hour glucose concentrations. In both patient populations, the treatment proved safe for use and well tolerated at the tested regimen.